A1-026

IN VIVO PK/PD OF MECILLINAM IN A MURINE URINARY TRACT INFECTION (UTI) MODEL

Vingsbo Lundberg C[1], Kvist P[2], Kemp K[2], Jensen K.S[1], Frimodt-Møller N[1]
1) Statens Serum Institut, Copenhagen, Denmark 2) LEO Pharma, Ballerup, Denmark

Carina Vingsbo Lundberg
CVL@ssi.dk
National Center for Antimicrobials and Infection Control
5, Artillerivej, DK-2300
Copenhagen S
Denmark

STATENS SERUM INSTITUT

National Center for Antimicrobials and Infection Control

BACKGROUND

Mecillinam, MEC, (amdinocillin) is a promising drug for treatment of urinary tract infection (UTI) caused by ESBL-producing E.coli. We used an UTI model in mice to identify the PK/PD index that best correlates with efficacy of MEC.

MATERIALS AND METHODS

PK- Studies

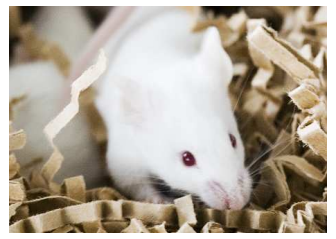

The PK characteristics of MEC was studied in OF-1 mice after single SC doses of 0.26 to 66.7 mg/kg. Blood and urine was collected from mice (n=3) starting at 6 min to 2 hrs after dosing and the drug levels were determined by MS.

Efficacy Studies

Urinary tract infection was induced via urethral catheterization with an ESBL-producing (CTX-M9) E.coli. Mice (n=8) were treated once, twice or four times daily at day 1 and 2 post infection. Urine, bladders and kidneys were collected day 3 post infection for CFU determination. Each dot in the figures above represents the mean of 8 mice.

PKPD modeling

Estimation of T>MIC, AUC_{24} and C_{max} for the various dosage regimens was done with the PKPDsim software package (1) based on the concentration-time points from the five primary single-bolus PK-studies.

References

1) PKPDsim software homepage, http://www.ssi.dk/pkpdsim

CONCLUSION

The murine experimental UTI model provides a means of studying PK/PD at clinically relevant sites of infection. Here we show that for MEC, the PD parameter that best correlated with efficacy was %T>MIC, and that very low values in blood, < 5% correlated with values of 33–46% in the urine.

Dose dependent Pharmacokinetics

Plasma and urine concentration of MEC after single dosing

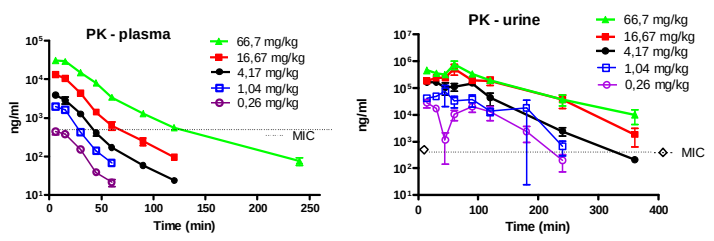
PK - plasma
PK - urine
66,7 mg/kg
16,67 mg/kg
4,17 mg/kg
1,04 mg/kg
0,26 mg/kg
MIC
ng/ml
Time (min)

T>MIC

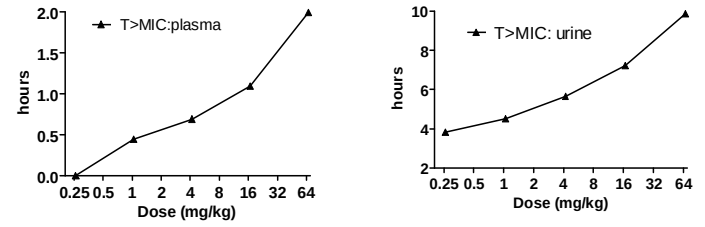
T>MIC:plasma
T>MIC: urine
hours
Dose (mg/kg)

Cpeak

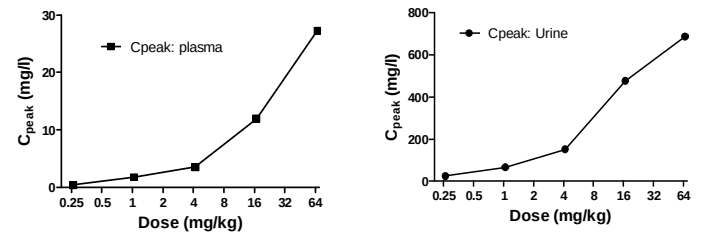
Cpeak: plasma
Cpeak: Urine
Cpeak (mg/l)
Dose (mg/kg)

AUC_{24}

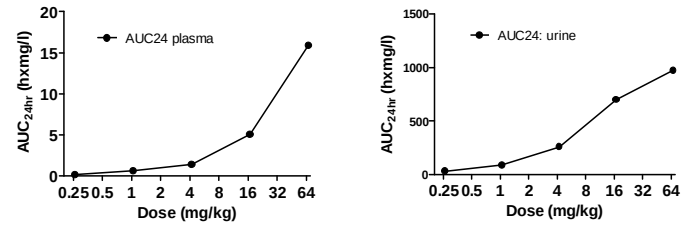
AUC24 plasma
AUC24: urine
AUC24hr (hxmg/l)
Dose (mg/kg)

Correlation of MEC PK-PD indices

in blood / **in urine**

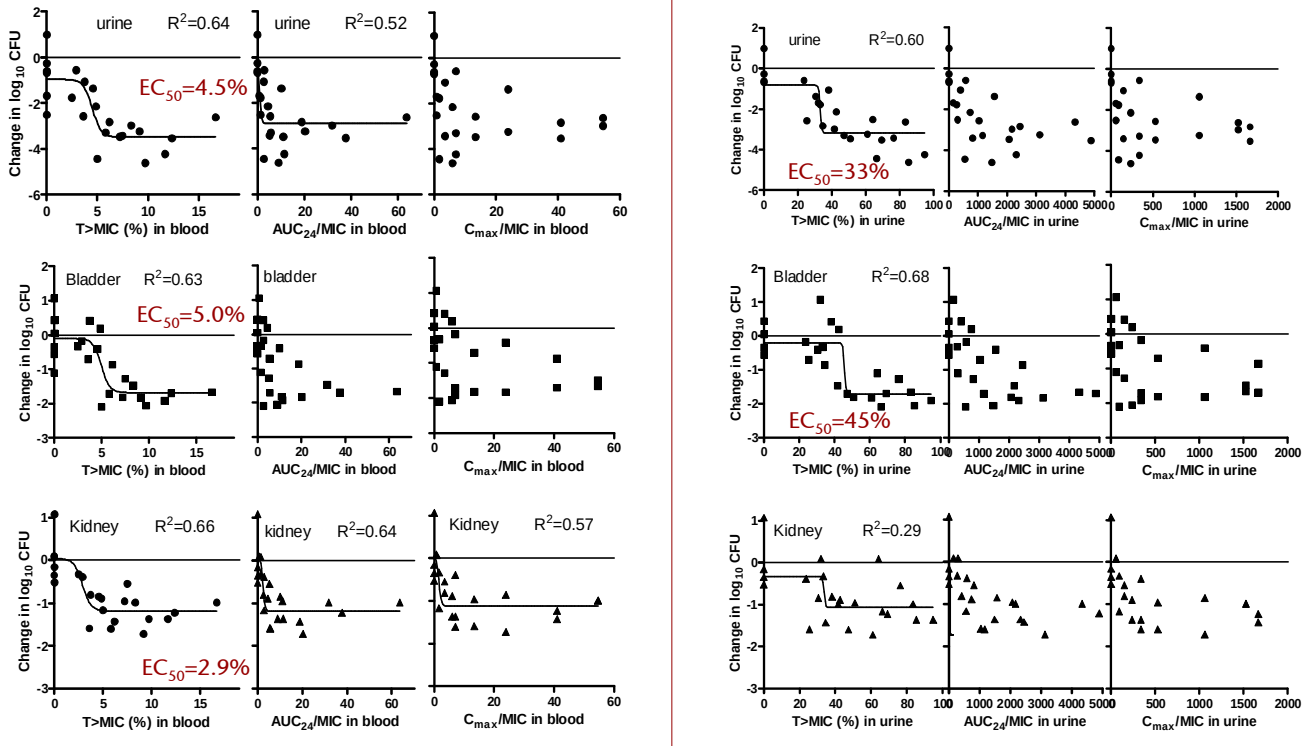
urine R2=0.64
EC50=4.5%
urine R2=0.52
Bladder R2=0.63
EC50=5.0%
bladder
Kidney R2=0.66
EC50=2.9%
kidney R2=0.64
Kidney R2=0.57
urine R2=0.60
EC50=33%
Bladder R2=0.68
EC50=45%
Kidney R2=0.29
Change in log10 CFU
T>MIC (%) in blood
AUC24/MIC in blood
Cmax/MIC in blood
T>MIC (%) in urine
AUC24/MIC in urine
Cmax/MIC in urine

Maximal effect on bacterial reduction:

- in urine was achieved at 7.9 % T>MIC in blood, corresponding to 37% T>MIC in urine
- in bladder was achieved at 7.2 % T>MIC in blood, corresponding to 48% T>MIC in urine
- in kidney was achieved at 5.5 % T>MIC in blood

Relationship between dosing interval and efficacy

Urine / bladder / kidney

Doses/day

x1

x2

x4

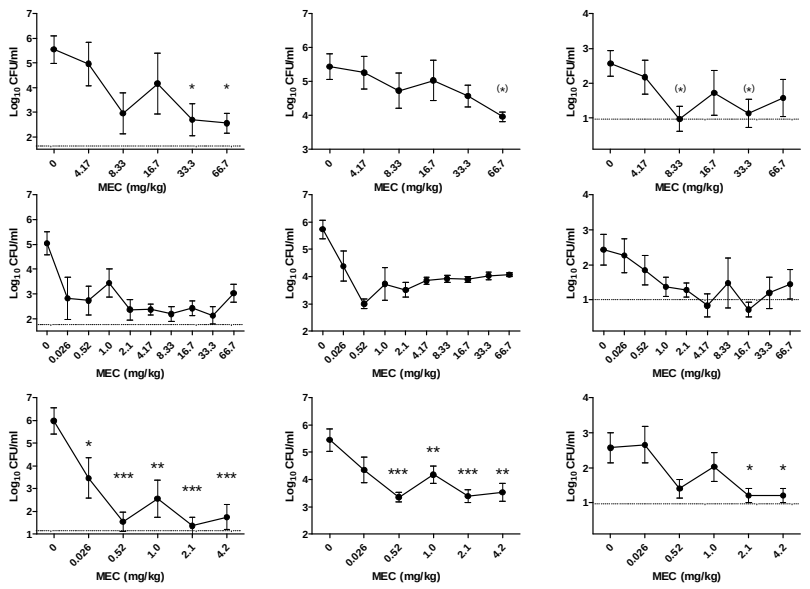
Log10 CFU/ml
MEC (mg/kg)

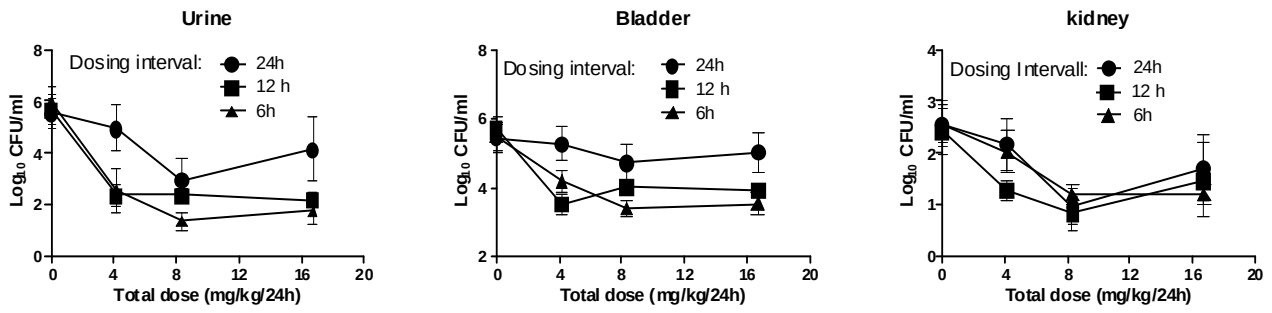
Urine
Bladder
kidney
Dosing interval:
Dosing Intervall:
24h
12 h
6h
Log10 CFU/ml
Total dose (mg/kg/24h)

Relationship between T>MIC and efficacy

%T>MIC in blood / **%T>MIC in urine**

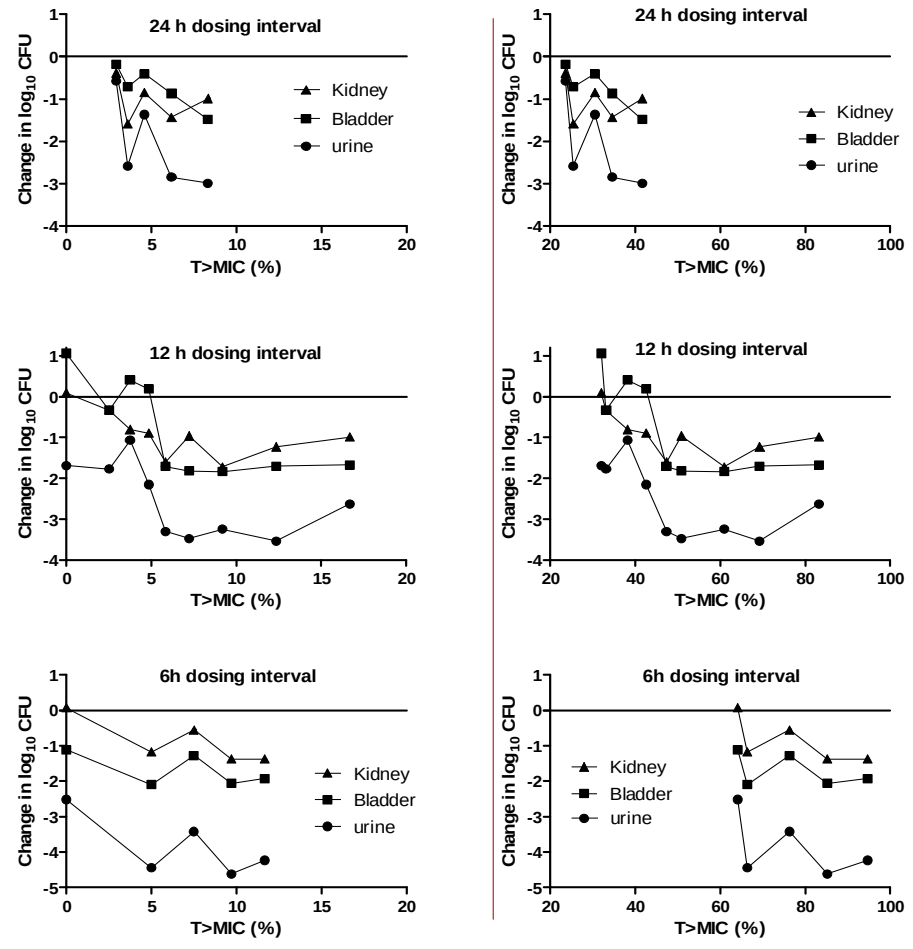
24 h dosing interval
12 h dosing interval
6h dosing interval
Kidney
Bladder
urine
Change in log10 CFU
T>MIC (%)